12 weeks safety and efficacy results of the novel C5 inhibitor coversin in PNH with resistance to eculizumab due to complement C5 polymorphism

Saskia Langemeijer[1], Wynne Weston-Davies[2],Miles A Nunn[2],Jun-Ichi Nishimura[3], Yuzuru Kanakura[3], Nicole Blijlevens[1], Marten Nijziel[1], Petra Muus[1]

[1]Department of Hematology, Radboudumc, Nijmegen, Netherlands; [2]Akari Therapeutics Plc, London, United Kingdom; [3]Department of Hematology and Oncology, Osaka University Graduate School of Medicine, Suita, Japan

Introduction

Paroxysmal Nocturnal Haemoglobinuria (PNH) is a rare acquired life-threatening disease characterized by complement induced haemolysis and its sequelae and a high incidence of thrombosis. The monoclonal antibody eculizumab binds to C5 and prevents its activation and cleavage into C5a and C5b and is an established treatment for PNH and aHUS. However, for patients with the rare amino acid polymorphism p.Arg885His or pArg885Ser, which interferes with the binding and efficacy of eculizumab (Nishimura et al, 2014, Langemeijer et al, 2015), there is still no effective treatment.

A new small protein complement inhibitor named Coversin is in Phase 1-2 clinical development. Coversin also prevents cleavage and activation of C5 but binds to C5 at a different site than eculizumab. In vitro Coversin inhibits C5 activation in both wild type C5 and C5 with a polymorphism at the Eculizumab binding site. Coversin studies in healthy volunteers proved safe and demonstrated inhibition of terminal complement activation.

We report safety and preliminary efficacy data of the novel C5 inhibitor Coversin in a severely haemolytic PNH patient with a C5 polymorphism.

Patient

The patient is a 30 year old male with PNH, (granulocyte clone size: 90%) and severe haemolysis (LDH 3 to 17 x upper limit of normal), transient renal failure, extreme fatigue and symptoms of muscle dystonia and no history of thrombosis. He remained severely haemolytic during eculizumab treatment despite adequate drug levels and no human antidrug antibodies. Other causes of haemolysis were excluded. The patient was shown to have a p.Arg885Ser polymorphism in the C5 gene rendering him resistant to eculizumab therapy (Figure 1)

Coversin was administered by s.c. injection at an ablating dose of 0.57 mg/kg on day 1, followed by a maintenance dose of 0.14 mg/kg per day thereafter. Peripheral blood samples were drawn for PK/PD. Protocol specified doubling of the dose and /or shortening of the dose interval were allowed on the basis of clinical symptoms and CH50 levels to achieve adequate and sustained complement inhibition.

Contact information

Saskia Langemeijer
Radboud University Medical Centre
Nijmegen, The Netherlands

Phone: +31 (0)24 36 14762
Fax: +31 (0)24 35 42080
E-mail: saskia.langemeijer@radboudumc.nl

Figure 1: Sequence analysis of exon 21 of C5 in index patient

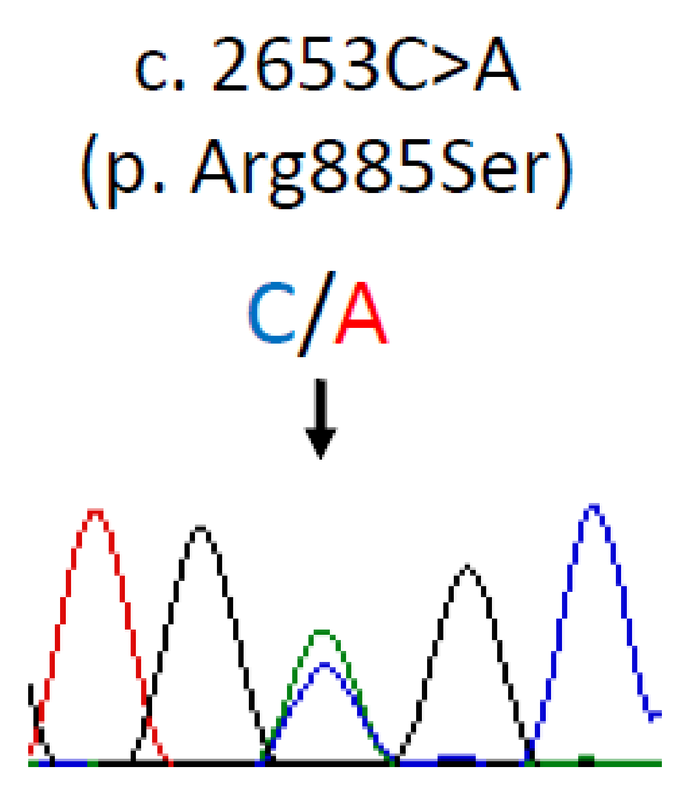
c. 2653C>A
(p. Arg885Ser)
C/A

DNA analysis of the coding region of C5 was performed in our index patient. This showed a single C5 heterozygous missense mutation, c.2653C>A, which predicts p.Arg885Ser . The same mutation was demonstrated in the DNA of our patient's healthy father.

Figure 2: LDH level and complement activity (CH50) during the first 17 days of treatment with Coversin

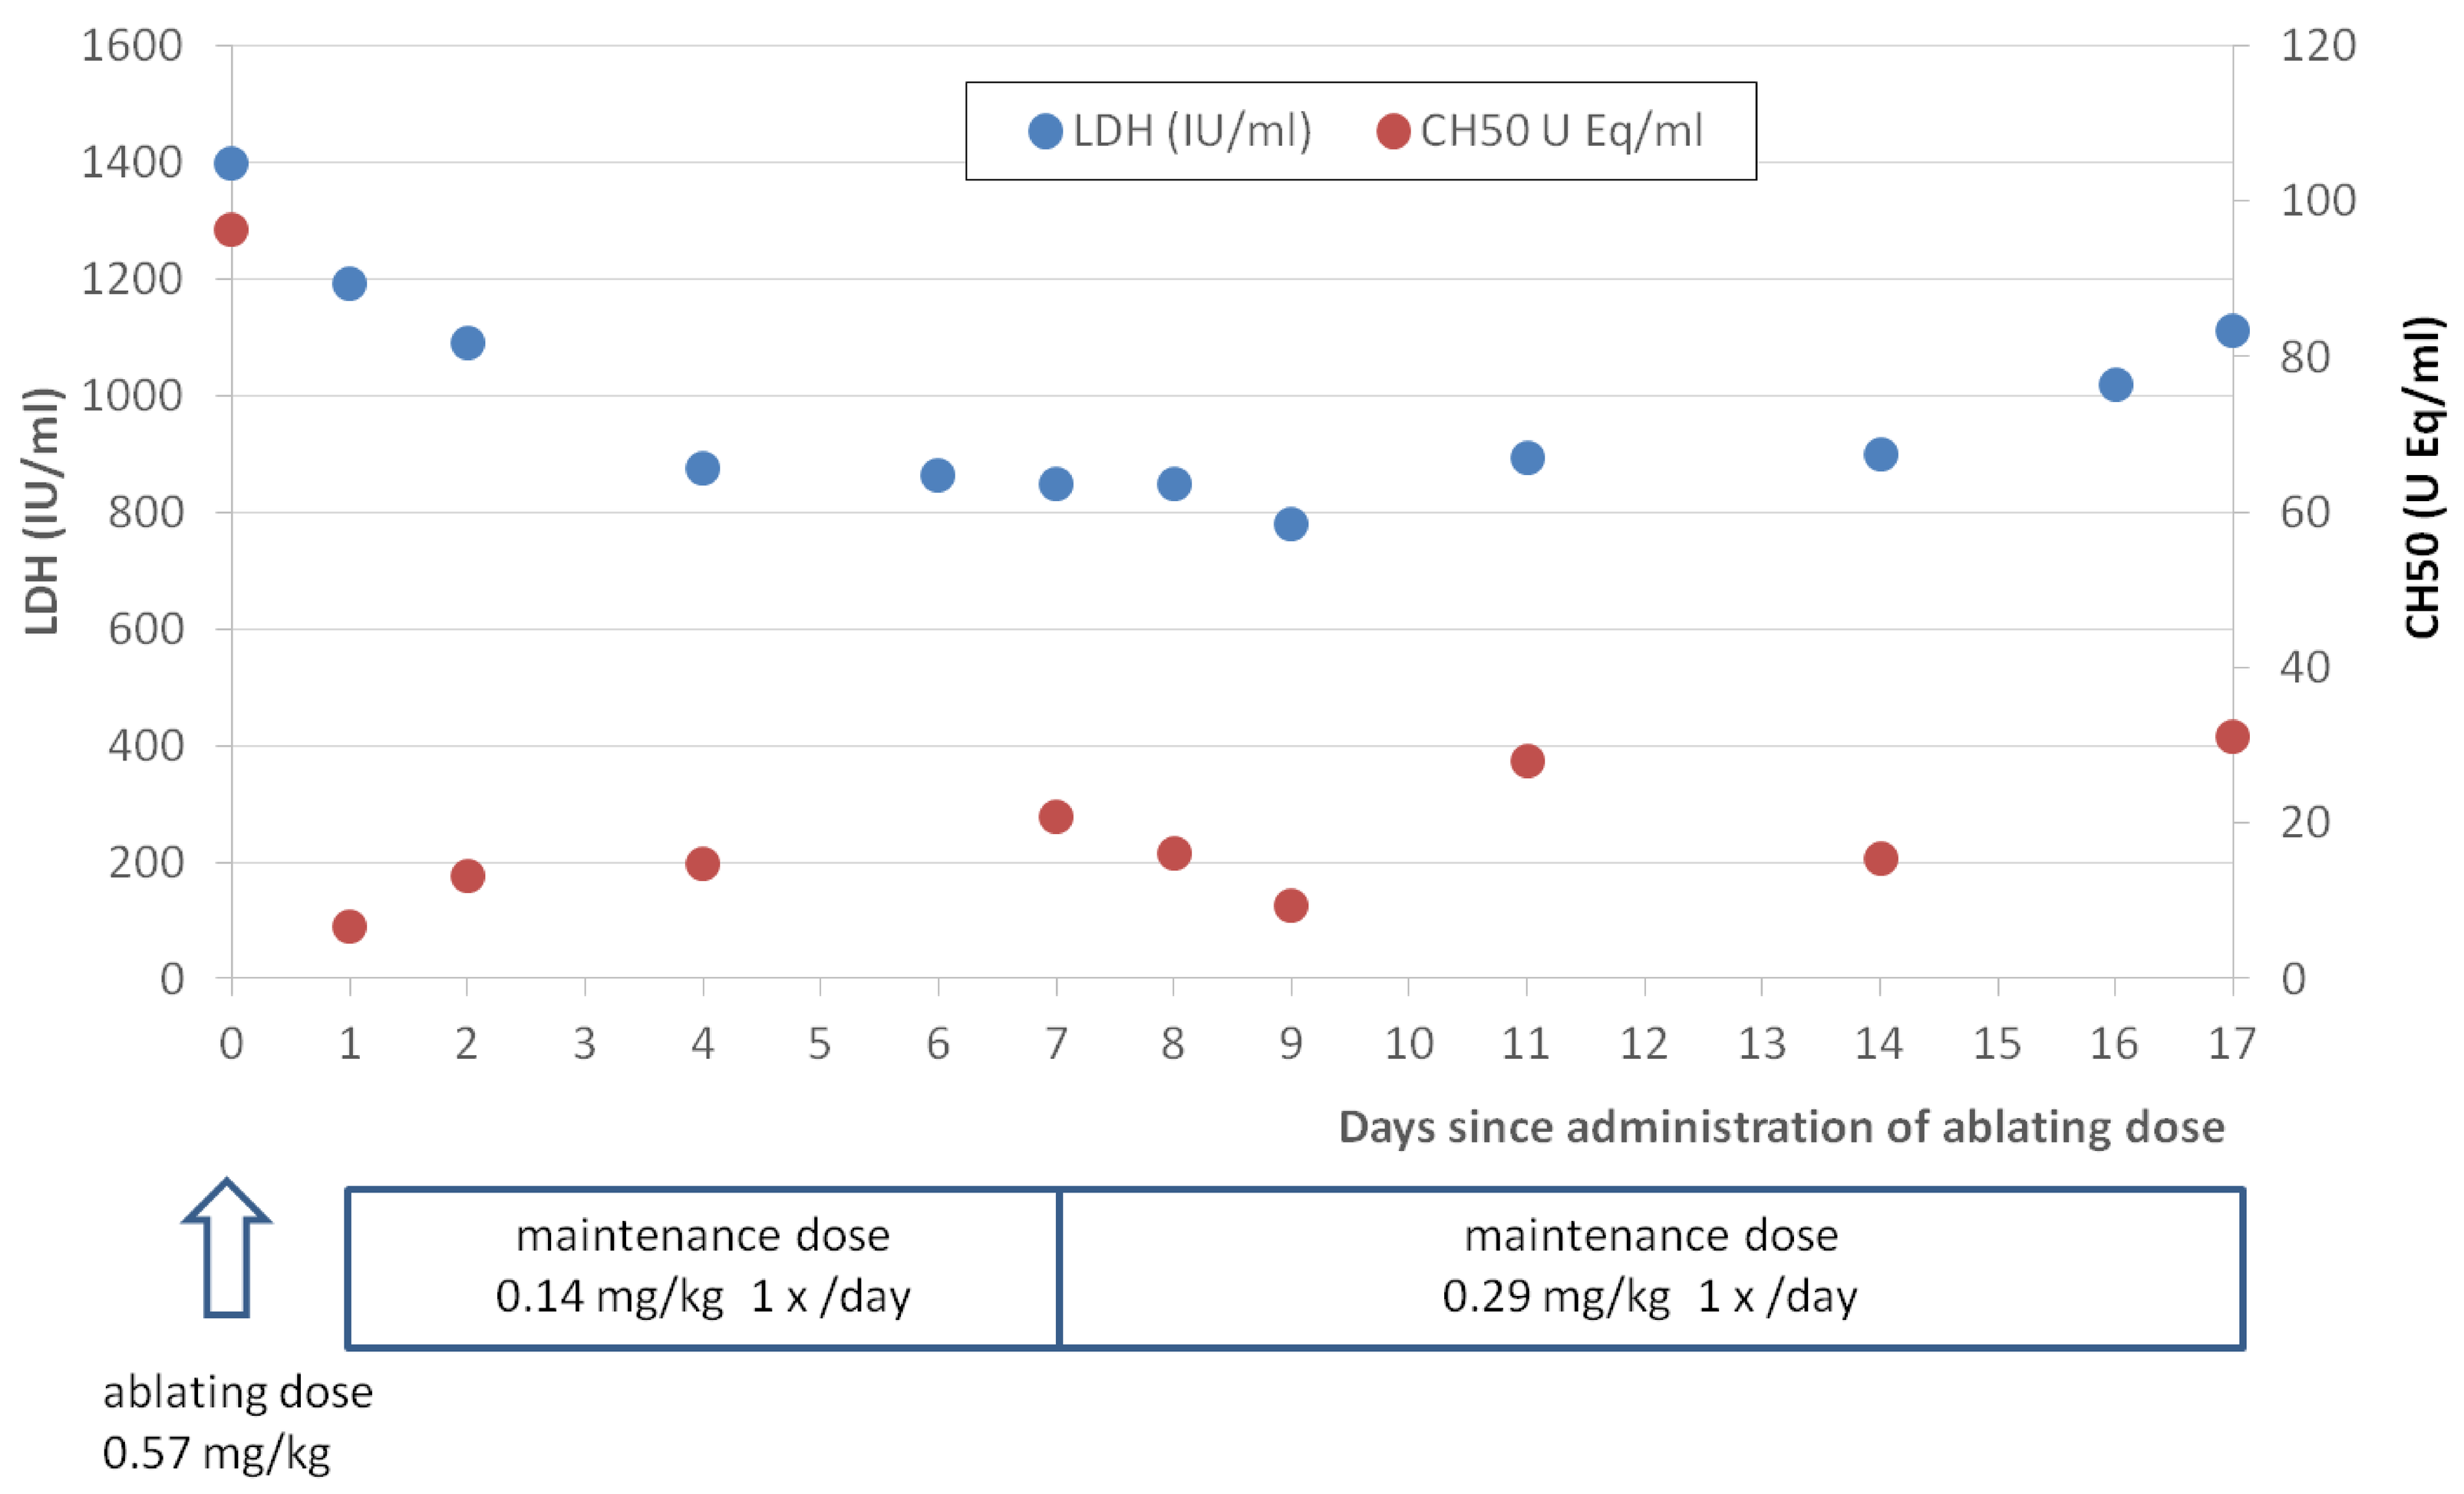
LDH (IU/ml)
CH50 U Eq/ml
LDH (IU/ml)
CH50 (U Eq/ml)
Days since administration of ablating dose
maintenance dose
0.14 mg/kg 1 x /day
maintenance dose
0.29 mg/kg 1 x /day
ablating dose
0.57 mg/kg

There was a good initial response to an ablating dose of 0.57mg/kg Coversin with CH50 levels decreasing below 8 U Eq/ml, which is the lower limit of qualification of the ELISA assay. Clinical symptoms and laboratory markers of haemolysis improved during the maintenance treatment of 0.14mg/kg every 24 hours. However, 6 days into the treatment, the patient again experienced haemolysis-associated symptoms with dark urine hours before the next s.c. injection and no further decrease of his LDH. The same occurred after doubling of the dose to 0.29 mg/kg per day.

Figure 3: LDH level and complement activity (CH50) after changing the dosing schedule to 0.14 mg/kg every 12 hours

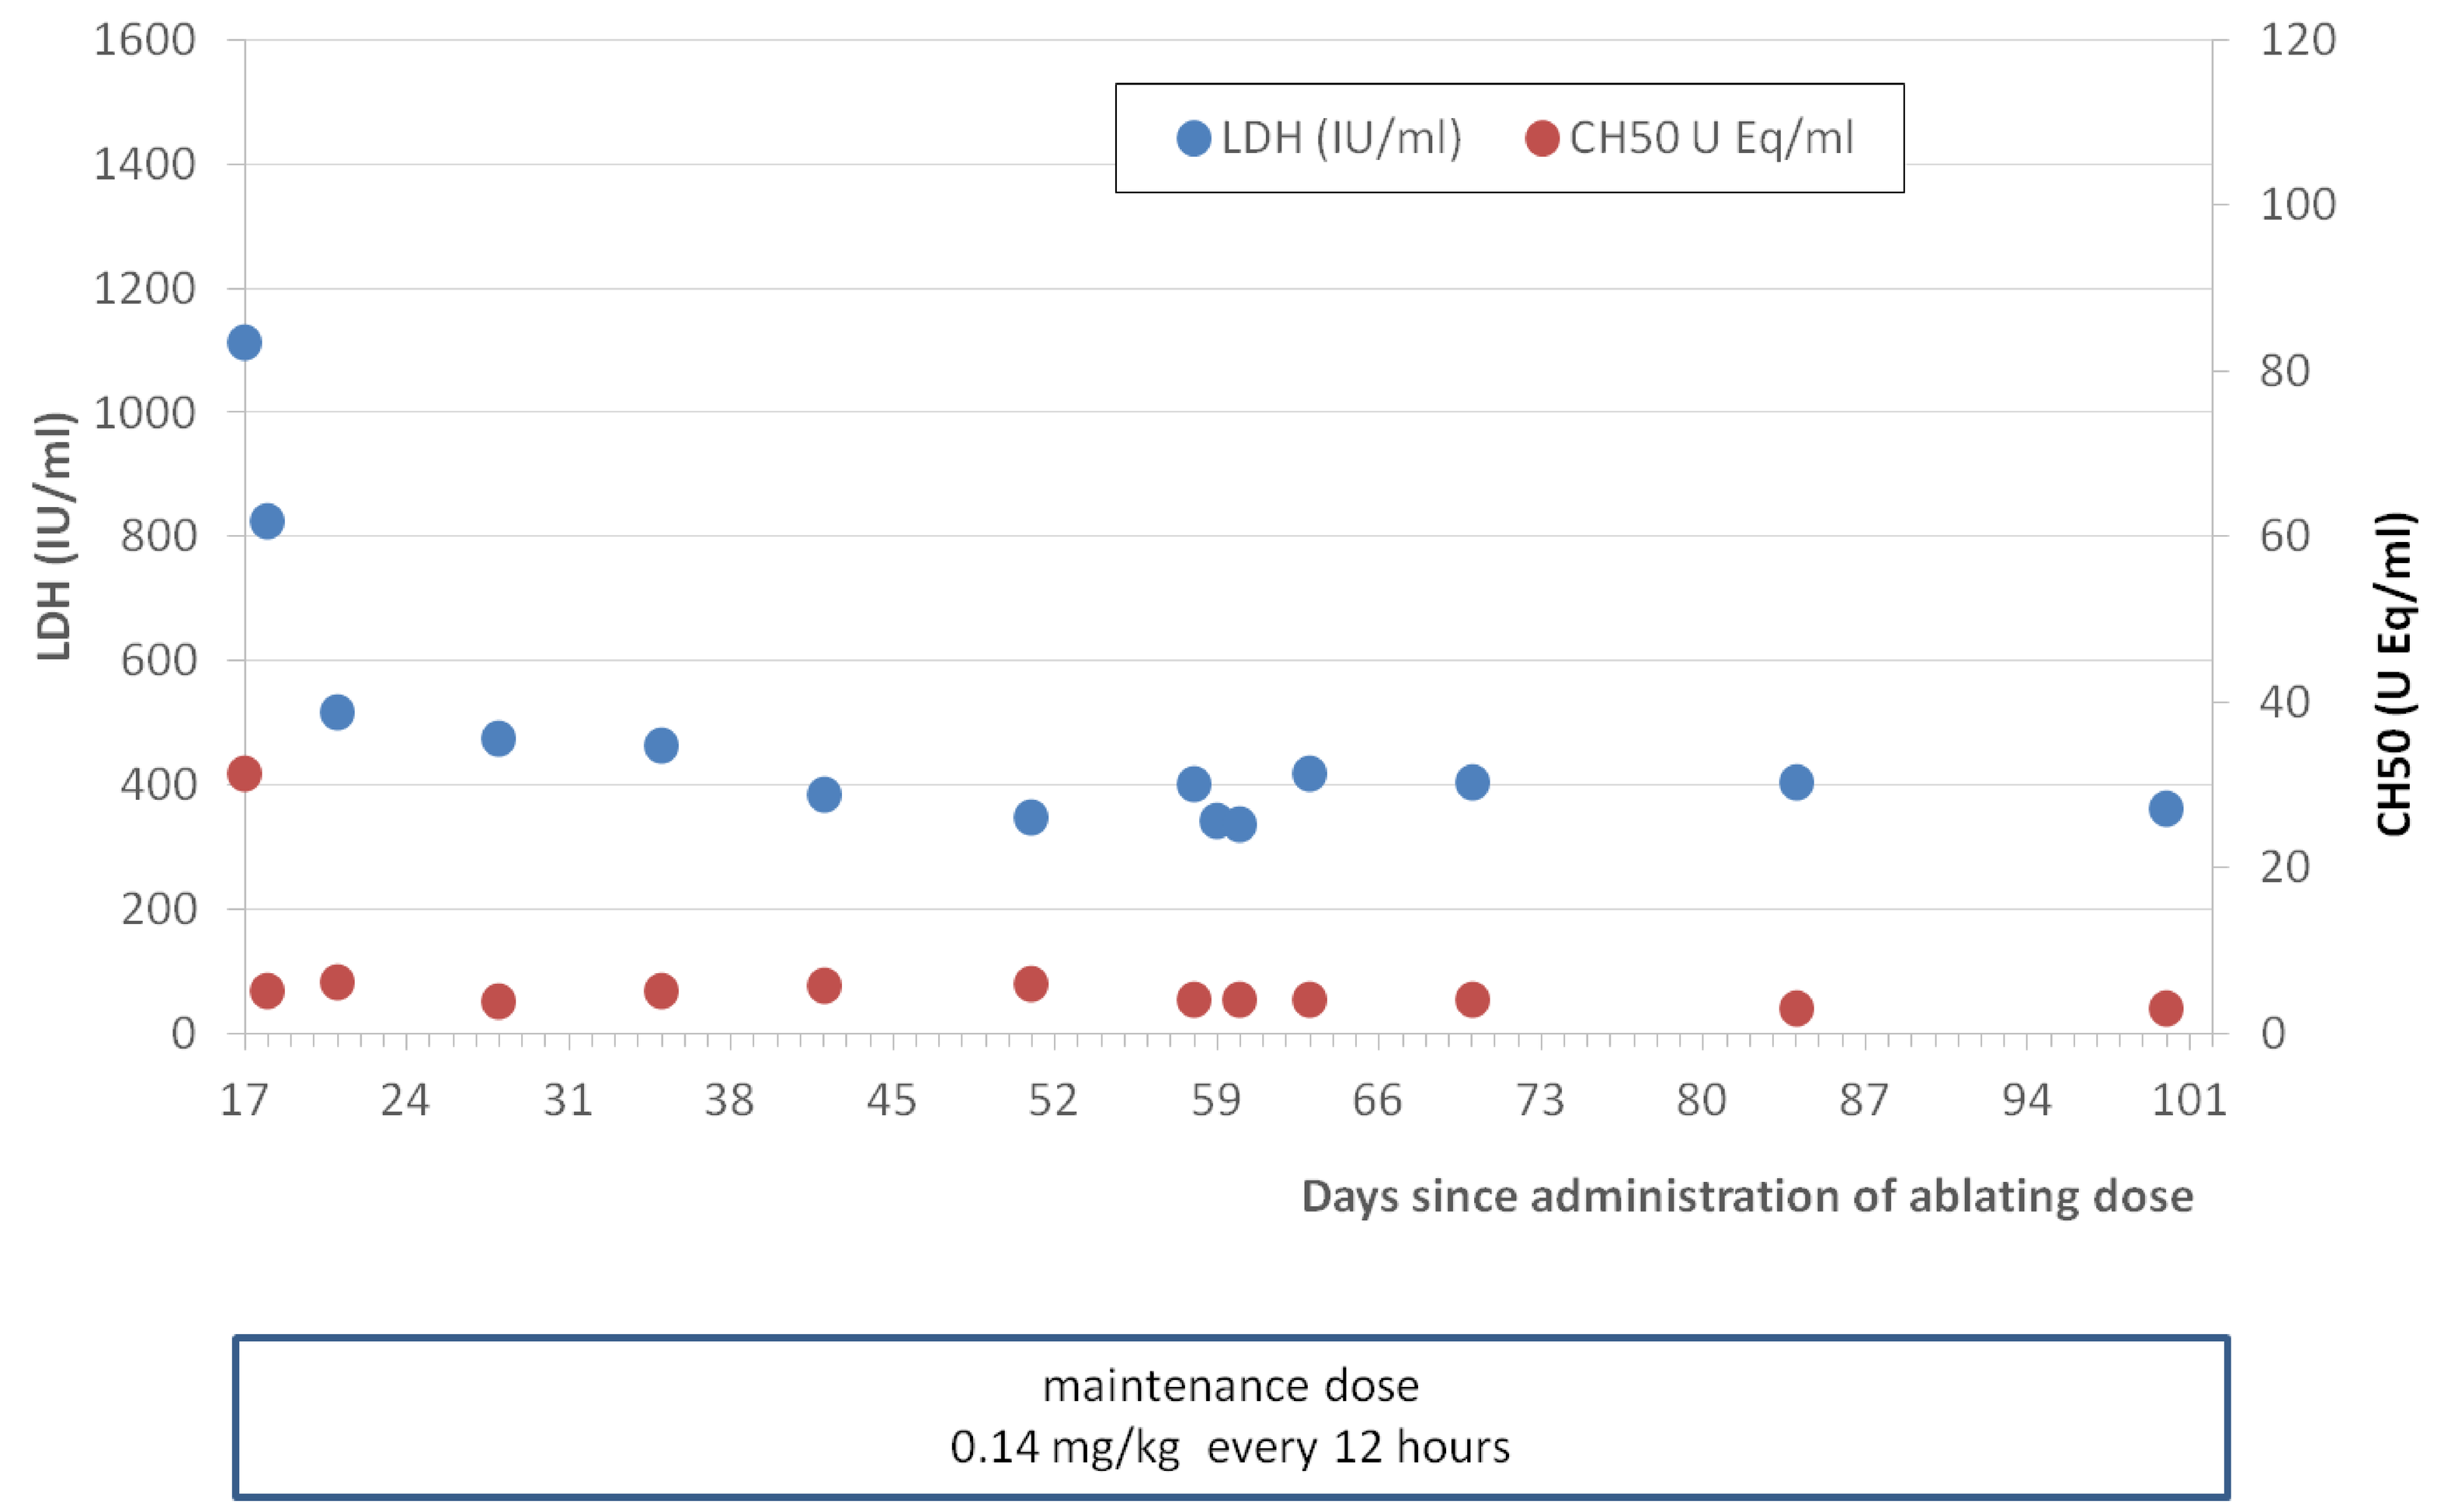
LDH (IU/ml)
CH50 U Eq/ml
LDH (IU/ml)
CH50 (U Eq/ml)
Days since administration of ablating dose
maintenance dose
0.14 mg/kg every 12 hours

Measurement of CH50 activity at 0,3,6,9,12,18 and 24 hours after administration of Coverin (data not shown) showed that, although a dosage of 0.14 or 0.29 mg/kg was sufficient to suppress complement activity, complement activity started to increase after 12 hours. Dividing the dose into 0.14 mg/kg injections Q 12 hours resulted in stable complement inhibition with CH50 levels <8 U Eq/ml and no breakthrough symptoms (Figure 3). The LDH decreased to approximately 1.5xUNL.

For the first time since he was diagnosed with PNH in 2009, the patient feels well with no symptoms of fatigue and no muscle dystonia. There have been no further haemolytic episodes. Twelve weeks after start of treatment, our patient did not have any drug-related adverse events, except occasional local and transient irritation at the injection site.

Conclusions

- Over a period of 12 weeks Coversin has proven safe and effective in the first PNH patient treated with the drug.

- Coversin may prove a useful alternative to Eculizumab for patients with resistance due to C5 polymorphisms.

Radboudumc